755-P
ADA 2023

A phase 2 evaluation of a novel GLP-1 analog ecnoglutide (XW003) for glycemic control in adults with type 2 diabetes

Dalong Zhu[1], Hong Qin[2], Qing Zheng[2], Jing Ning[2], Zhiyi Zhu[2], Mengying Guo[2], Yue Bu[2], Catherine L. Jones[3], Martijn Fenaux[3], Susan Xu[3], Mohammed K. Junaidi[3]
[1]Nanjing Drum Tower Hospital, Nanjing, China [2]Hangzhou Sciwind Biosciences, Hangzhou, China [3]Sciwind Biosciences, San Ramon, USA

BACKGROUND

Type 2 diabetes mellitus (T2DM) is an impactful global health problem, with approximately 483 million adults currently living with the disease. Diabetes can lead to serious complications, including retinopathy and neuropathy, as well as increase the risk for cardiovascular-related morbidity and mortality[1].

Ecnoglutide (XW003) is a novel, long-acting, glucagon-like peptide-1 (GLP-1) analog being developed for the treatment of T2DM and obesity. GLP-1 analogs mimic the activity of the natural incretin hormone, which is released after a meal. GLP-1 increases insulin production, lowers glucagon secretion, slows gastric emptying, and suppresses appetite. It acts in a glucose-dependent manner to lower blood sugar levels, maintaining homeostasis[2].

METHODS

We performed a randomized, double-blind, placebo controlled, Phase 2 clinical trial of ecnoglutide once weekly injection with 144 planned participants at 21 sites in China. Eligible male and female participants were 18-65 years of age, inclusive, had a diagnosis of T2DM[3] that was inadequately controlled by lifestyle or a single oral hypoglycemic agent. HbA1c was to be ≥7.5 to ≤10.5% and body mass index (BMI) between ≥20.0 to ≤35.0 kg/m² at screening.

Participants were randomized (1:1:1:1) to receive target doses of 0.4, 0.8, or 1.2 mg ecnoglutide or placebo as once weekly subcutaneous (SC) injections for 20 weeks, including a dose titration period. Participants were followed for 5 weeks after the end of treatment.

The primary efficacy endpoint was mean change from baseline in HbA1c levels at Week 20 (Day 134). Secondary efficacy endpoints included mean change from baseline in fasting plasma glucose (FPG), self-monitored blood glucose (SMBG), and body weight.

Study design

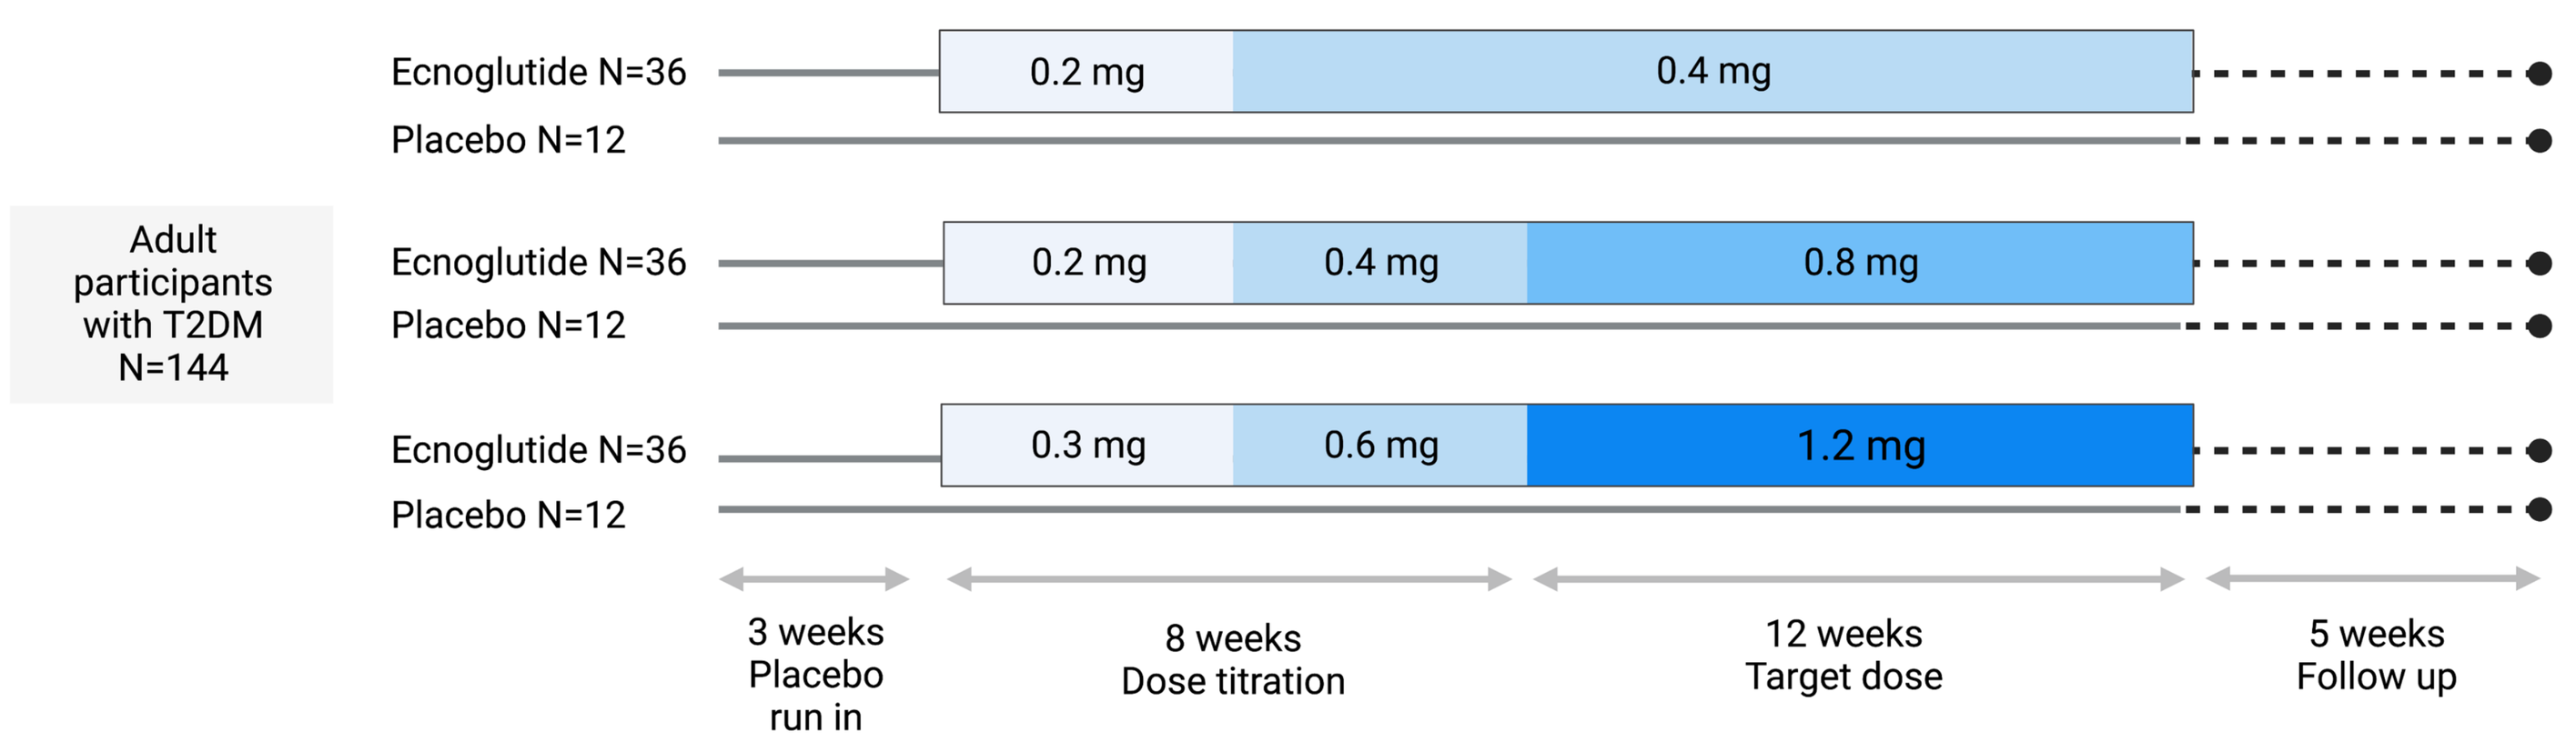
Adult participants with T2DM N=144
Ecnoglutide N=36
Placebo N=12
0.2 mg
0.4 mg
Ecnoglutide N=36
Placebo N=12
0.2 mg
0.4 mg
0.8 mg
Ecnoglutide N=36
Placebo N=12
0.3 mg
0.6 mg
1.2 mg
3 weeks Placebo run in
8 weeks Dose titration
12 weeks Target dose
5 weeks Follow up

Study number SCW0502-1021

RESULTS

Subject disposition

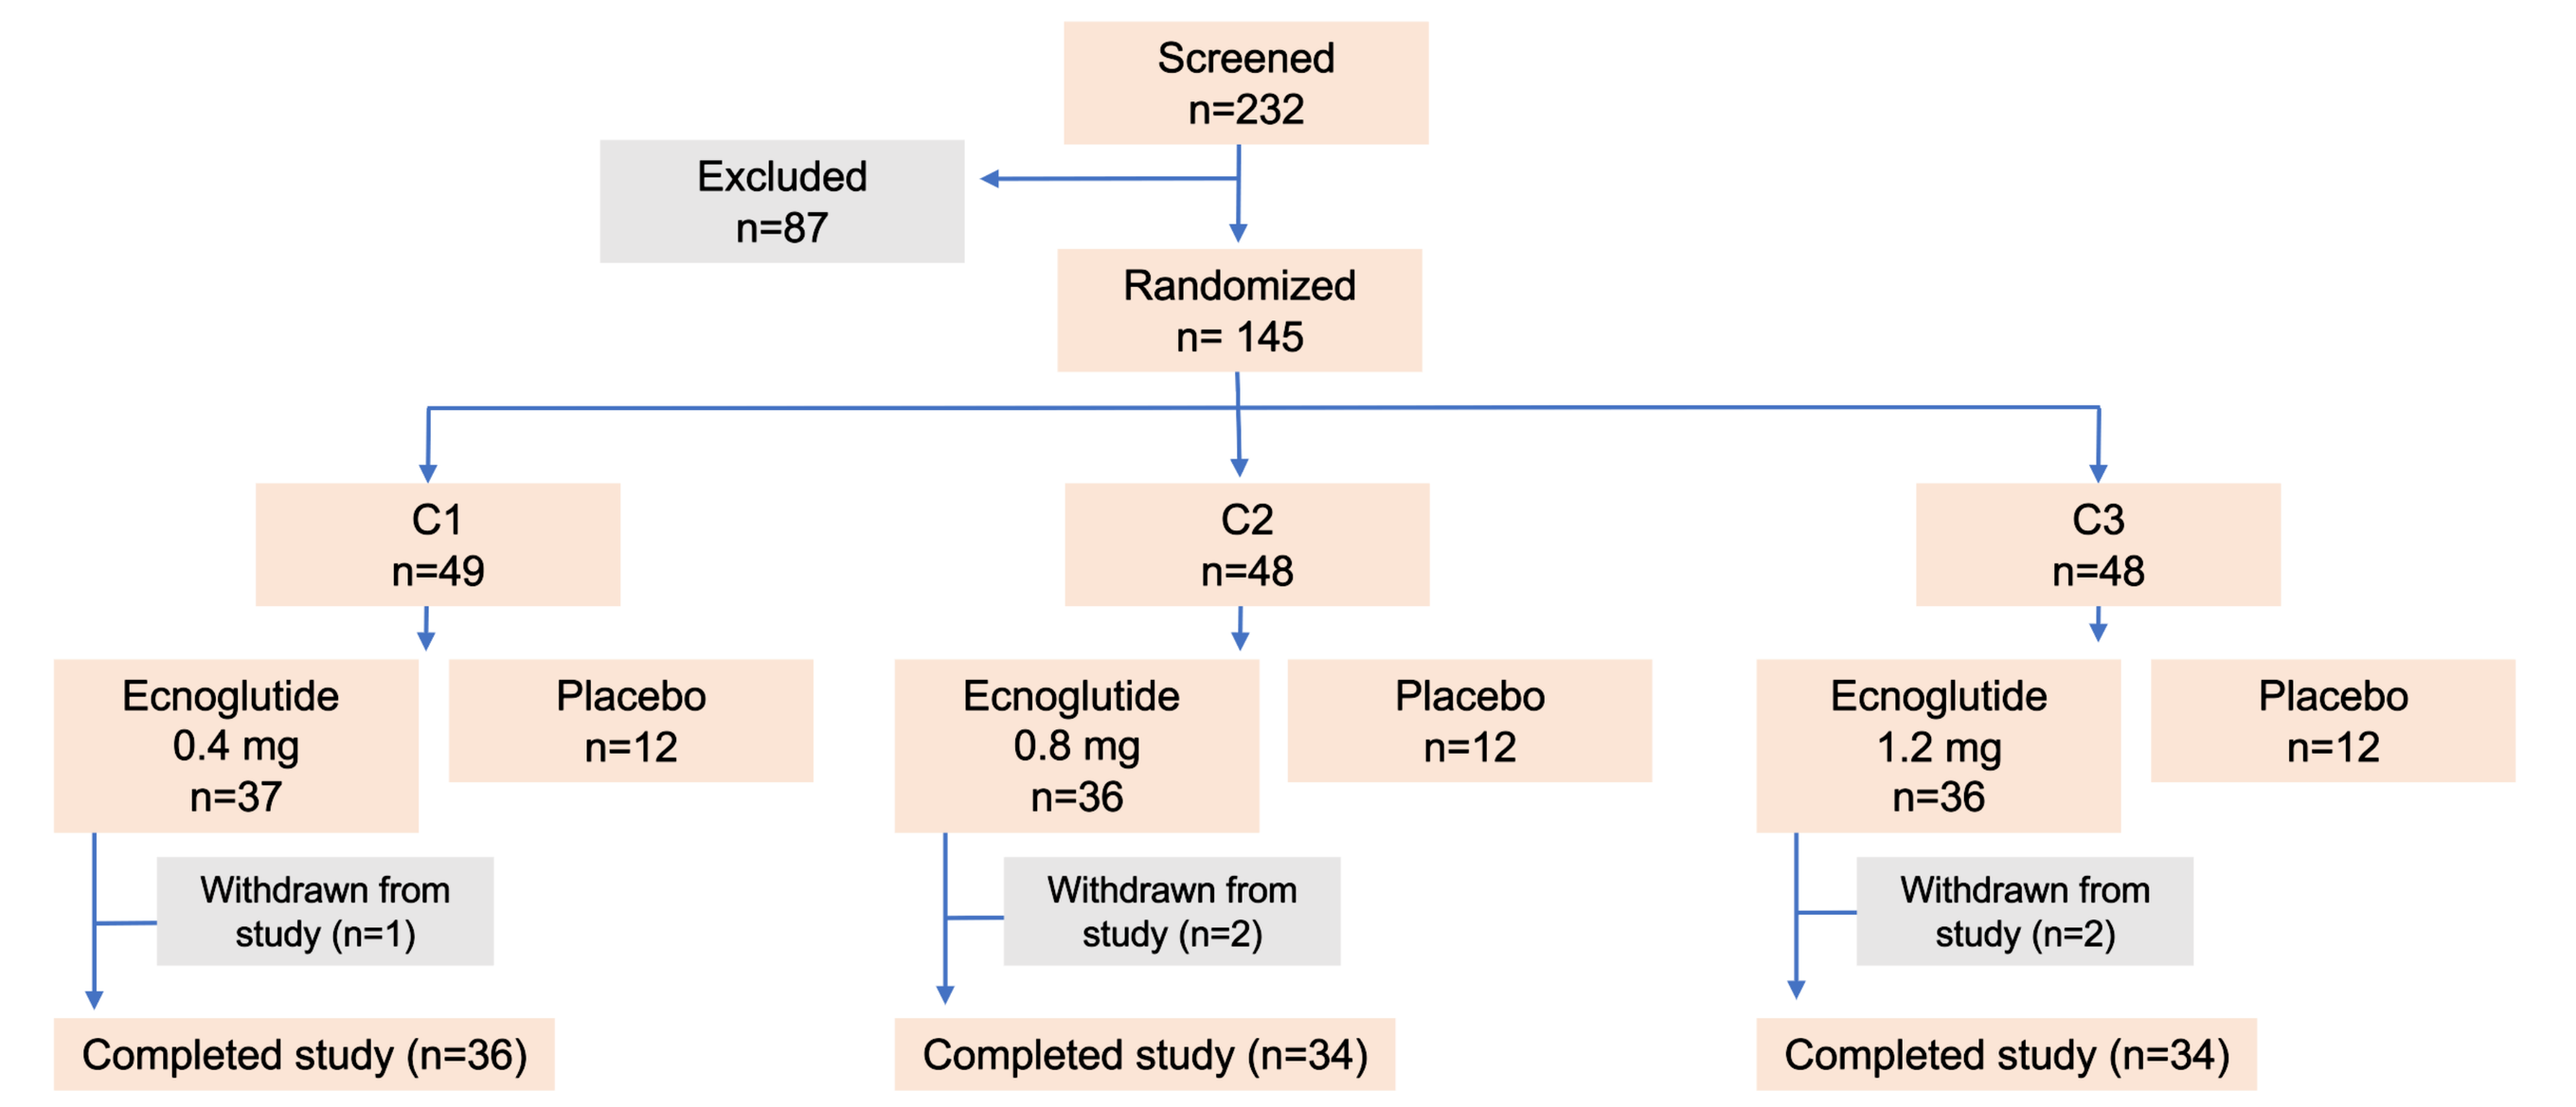
Screened n=232
Excluded n=87
Randomized n= 145
C1 n=49
C2 n=48
C3 n=48
Ecnoglutide 0.4 mg n=37
Placebo n=12
Ecnoglutide 0.8 mg n=36
Placebo n=12
Ecnoglutide 1.2 mg n=36
Placebo n=12
Withdrawn from study (n=1)
Withdrawn from study (n=2)
Withdrawn from study (n=2)
Completed study (n=36)
Completed study (n=34)
Completed study (n=34)

Overall, 109 participants received at least one dose of ecnoglutide and 104 (95.4%) completed the 20-week treatment period.

Demographics and baseline characteristics

Characteristic*	Ecnoglutide 0.4 mg (N=37)	Ecnoglutide 0.8 mg (N=36)	Ecnoglutide 1.2 mg (N=36)	Placebo (N=36)
Age (years)	49.1 (8.87)	51.8 (10.34)	49.6 (9.65)	49.7 (10.54)
Gender, n (%)				
Male	25 (67.6)	28 (77.8)	21 (58.3)	19 (52.8)
Female	12 (32.4)	8 (22.2)	15 (41.7)	17 (47.2)
Body weight (kg)	72.8 (14.6)	76.6 (13.4)	72.5 (12.1)	71.8 (12.2)
BMI (kg/m²)	25.8 (3.7)	26.6 (3.4)	26.2 (3.0)	26.4 (3.2)
HbA1c (%)	8.45 (0.64)	8.65 (0.76)	8.67 (0.75)	8.44 (0.67)
FPG (mmol/L)	9.70 (1.605)	11.08 (1.678)	10.00 (1.889)	10.50 (1.800)
Diabetes duration (months)	46.23 (51.93)	57.64 (44.18)	45.39 (49.55)	40.36 (35.58)
Previous antihyperglycemic treatment, n (%)	15 (40.5)	23 (63.9)	22 (61.1)	19 (52.8)

*Mean (SD) unless otherwise noted

HbA1c change from baseline

Ecnoglutide led to significant HbA1c reductions from baseline at end of treatment (P<0.0001 for all doses compared to placebo).

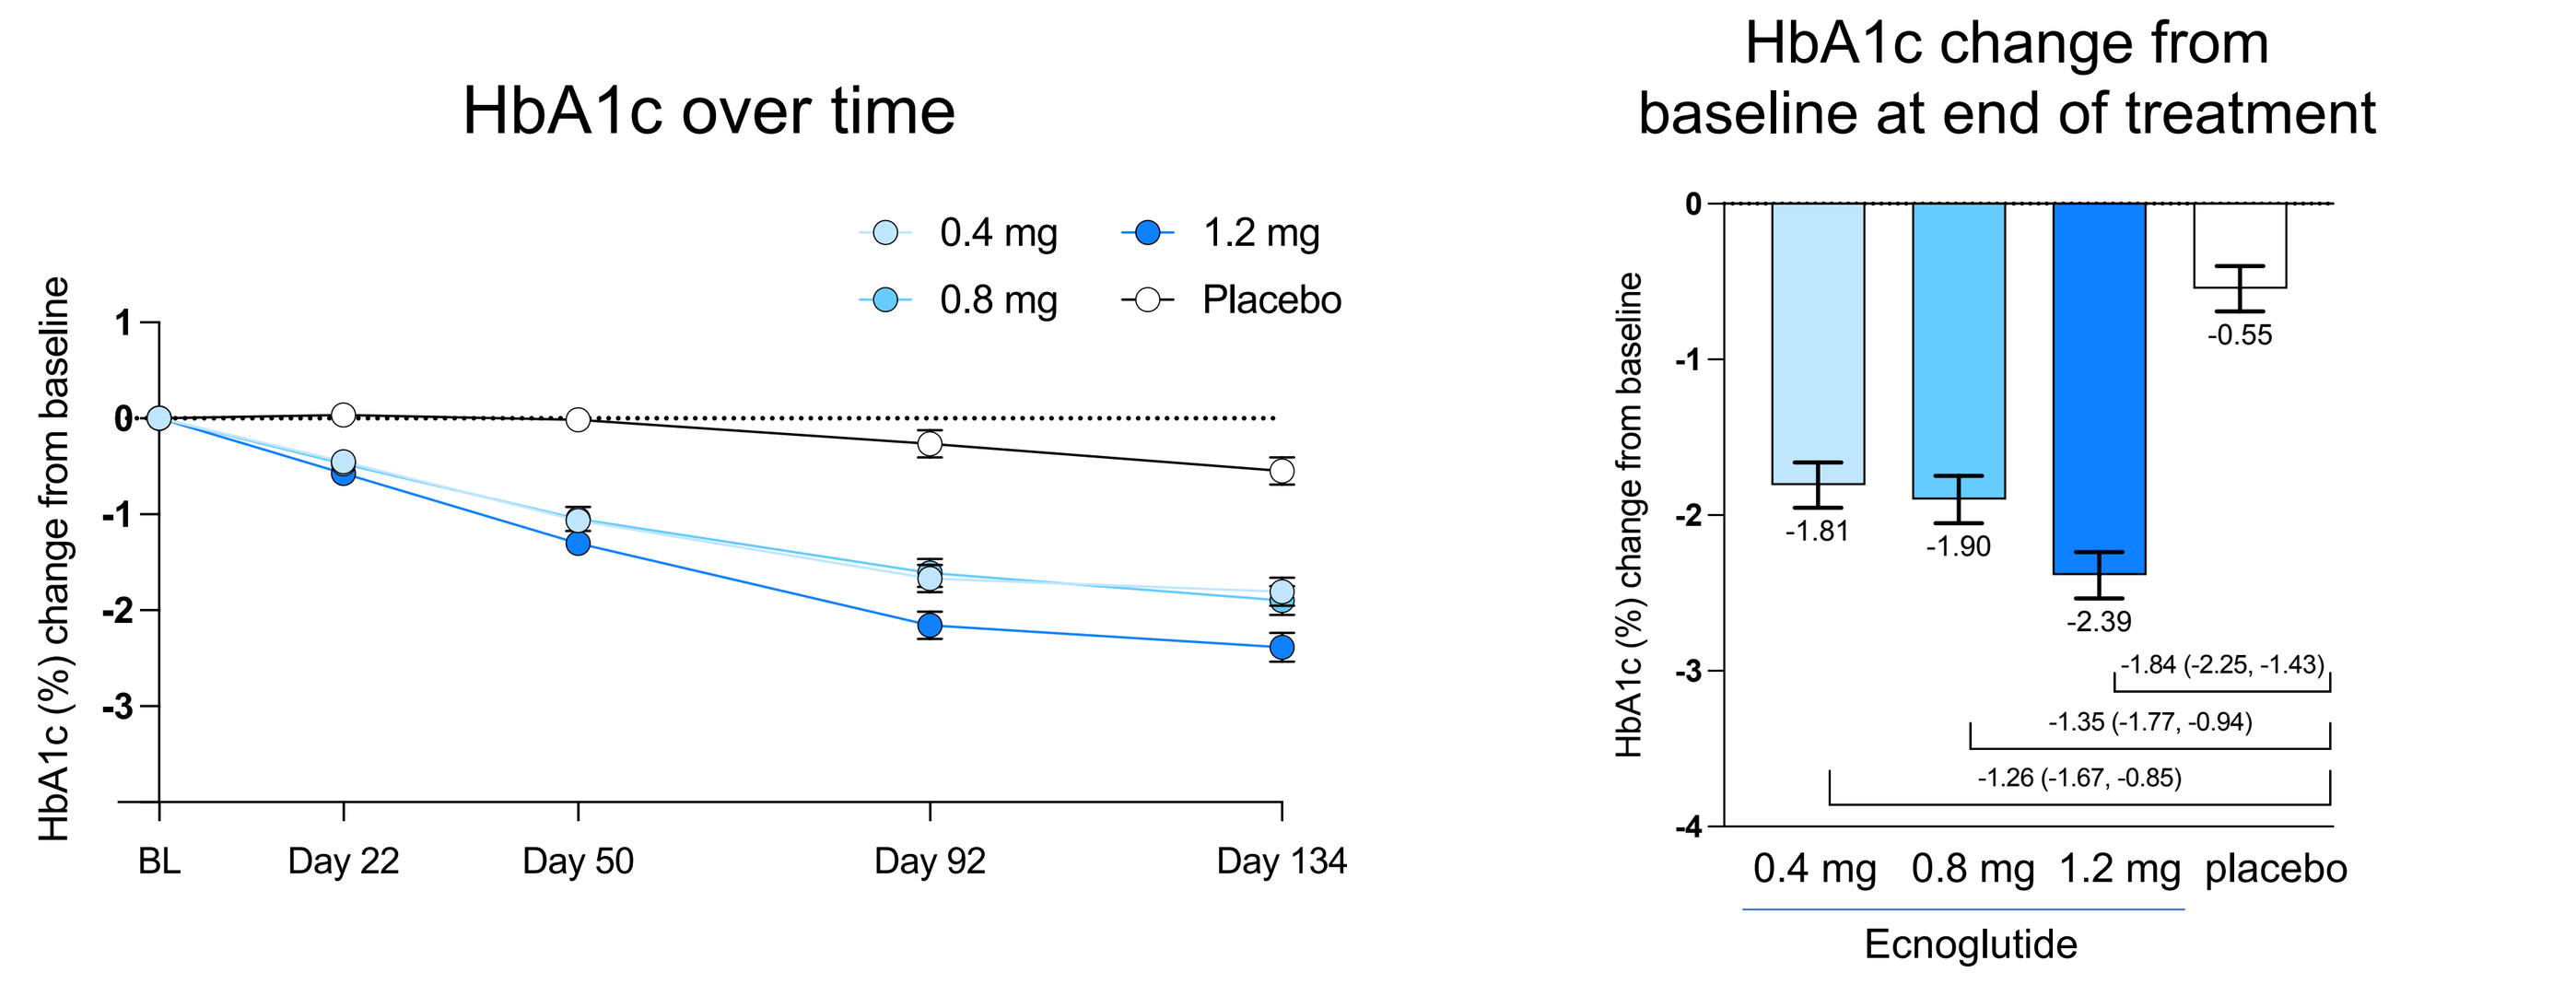
HbA1c over time
0.4 mg
0.8 mg
1.2 mg
Placebo
HbA1c (%) change from baseline
BL
Day 22
Day 50
Day 92
Day 134
HbA1c change from baseline at end of treatment
-1.81
-1.90
-2.39
-0.55
-1.84 (-2.25, -1.43)
-1.35 (-1.77, -0.94)
-1.26 (-1.67, -0.85)
0.4 mg
0.8 mg
1.2 mg
placebo
Ecnoglutide

HbA1c change from baseline over time and at end of treatment (least squares mean and standard error). Mean difference from placebo and 95% confidence interval are shown at end of treatment (Day 134).

A significantly higher proportion of participants receiving ecnoglutide reached HbA1c <7.0% and ≤6.5% at Week 20 vs placebo.

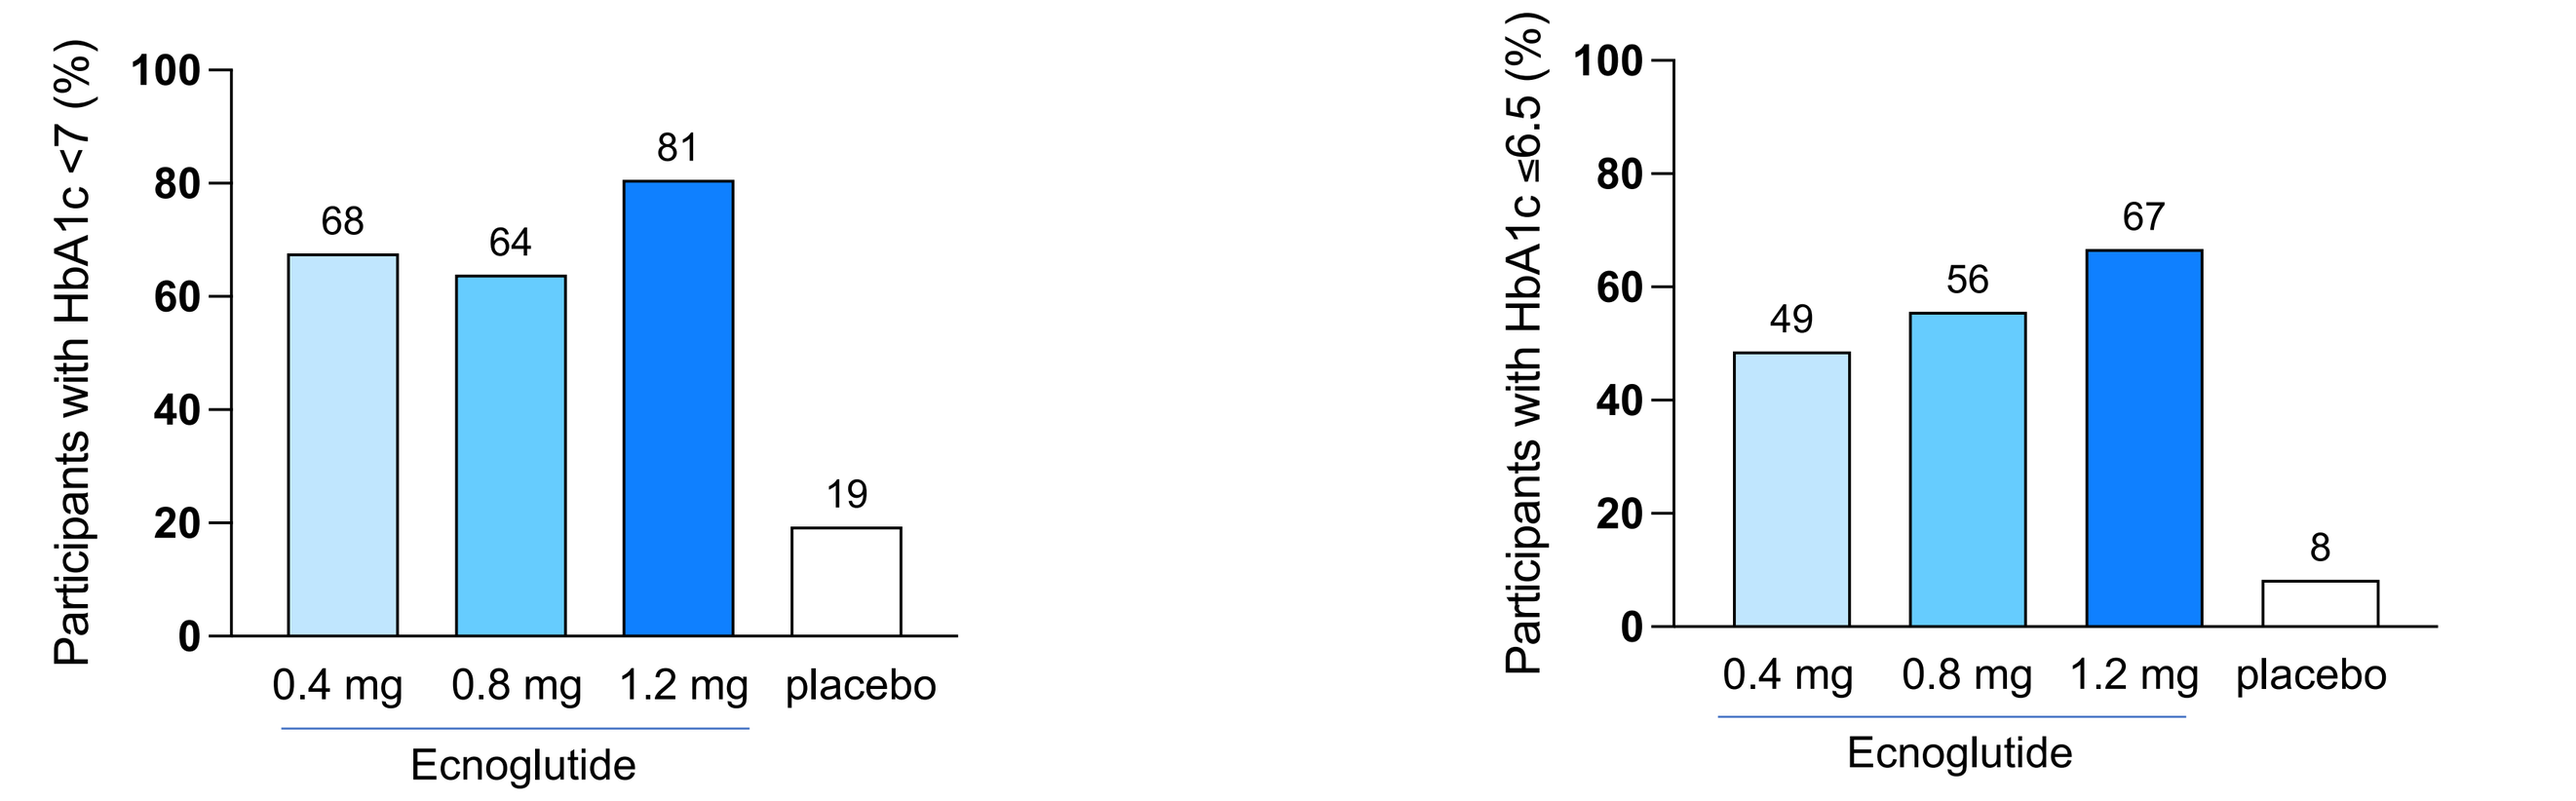
Participants with HbA1c <7 (%)
68
64
81
19
0.4 mg
0.8 mg
1.2 mg
placebo
Ecnoglutide
Participants with HbA1c ≤6.5 (%)
49
56
67
8
0.4 mg
0.8 mg
1.2 mg
placebo
Ecnoglutide

Proportion of participants with the American Diabetes Association (ADA) recommended HbA1c target of <7.0% and the American Association of Clinical Endocrinologists (AACE) target of ≤6.5% at Day 134, last observation carried forward (LOCF). P<0.001 for ecnoglutide versus placebo.

Blood glucose change from baseline

Ecnoglutide treatment led to significant decreases in fasting and average self-monitored blood sugar compared to placebo.

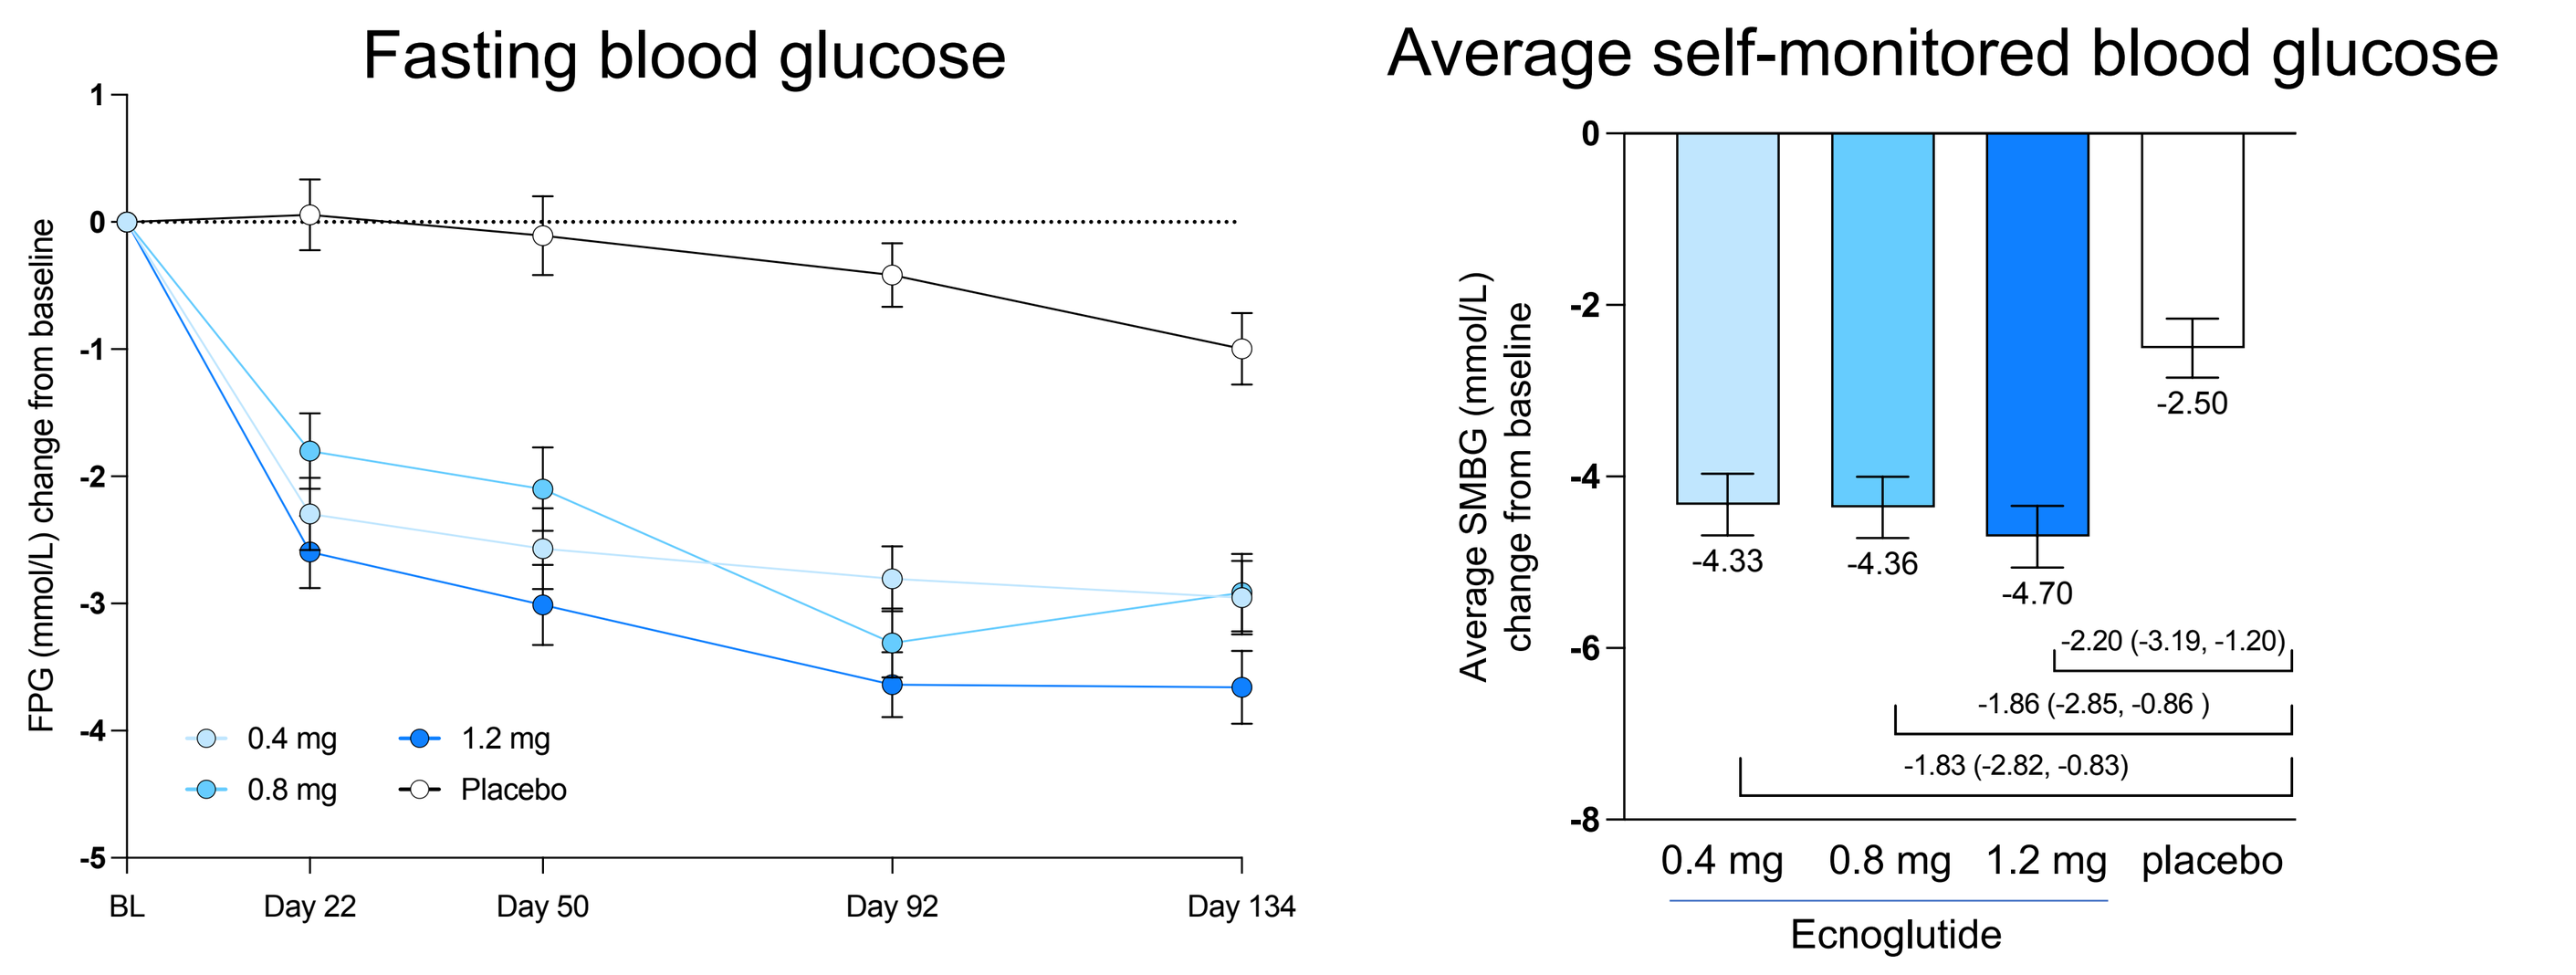
Fasting blood glucose
FPG (mmol/L) change from baseline
0.4 mg
0.8 mg
1.2 mg
Placebo
BL
Day 22
Day 50
Day 92
Day 134
Average self-monitored blood glucose
Average SMBG (mmol/L) change from baseline
-4.33
-4.36
-4.70
-2.50
-2.20 (-3.19, -1.20)
-1.86 (-2.85, -0.86)
-1.83 (-2.82, -0.83)
0.4 mg
0.8 mg
1.2 mg
placebo
Ecnoglutide

Least squares mean and standard error (SE) are shown, as well as mean difference from placebo and 95% confidence interval for SMBG. FPG, P<0.0001 for all ecnoglutide cohorts vs placebo at Days 22, 50, 92, and 134. SMBG, P<0.001 for all ecnoglutide cohorts compared to placebo.

Body weight change from baseline

Ecnoglutide treatment resulted in significant body weight reductions from baseline compared to placebo.

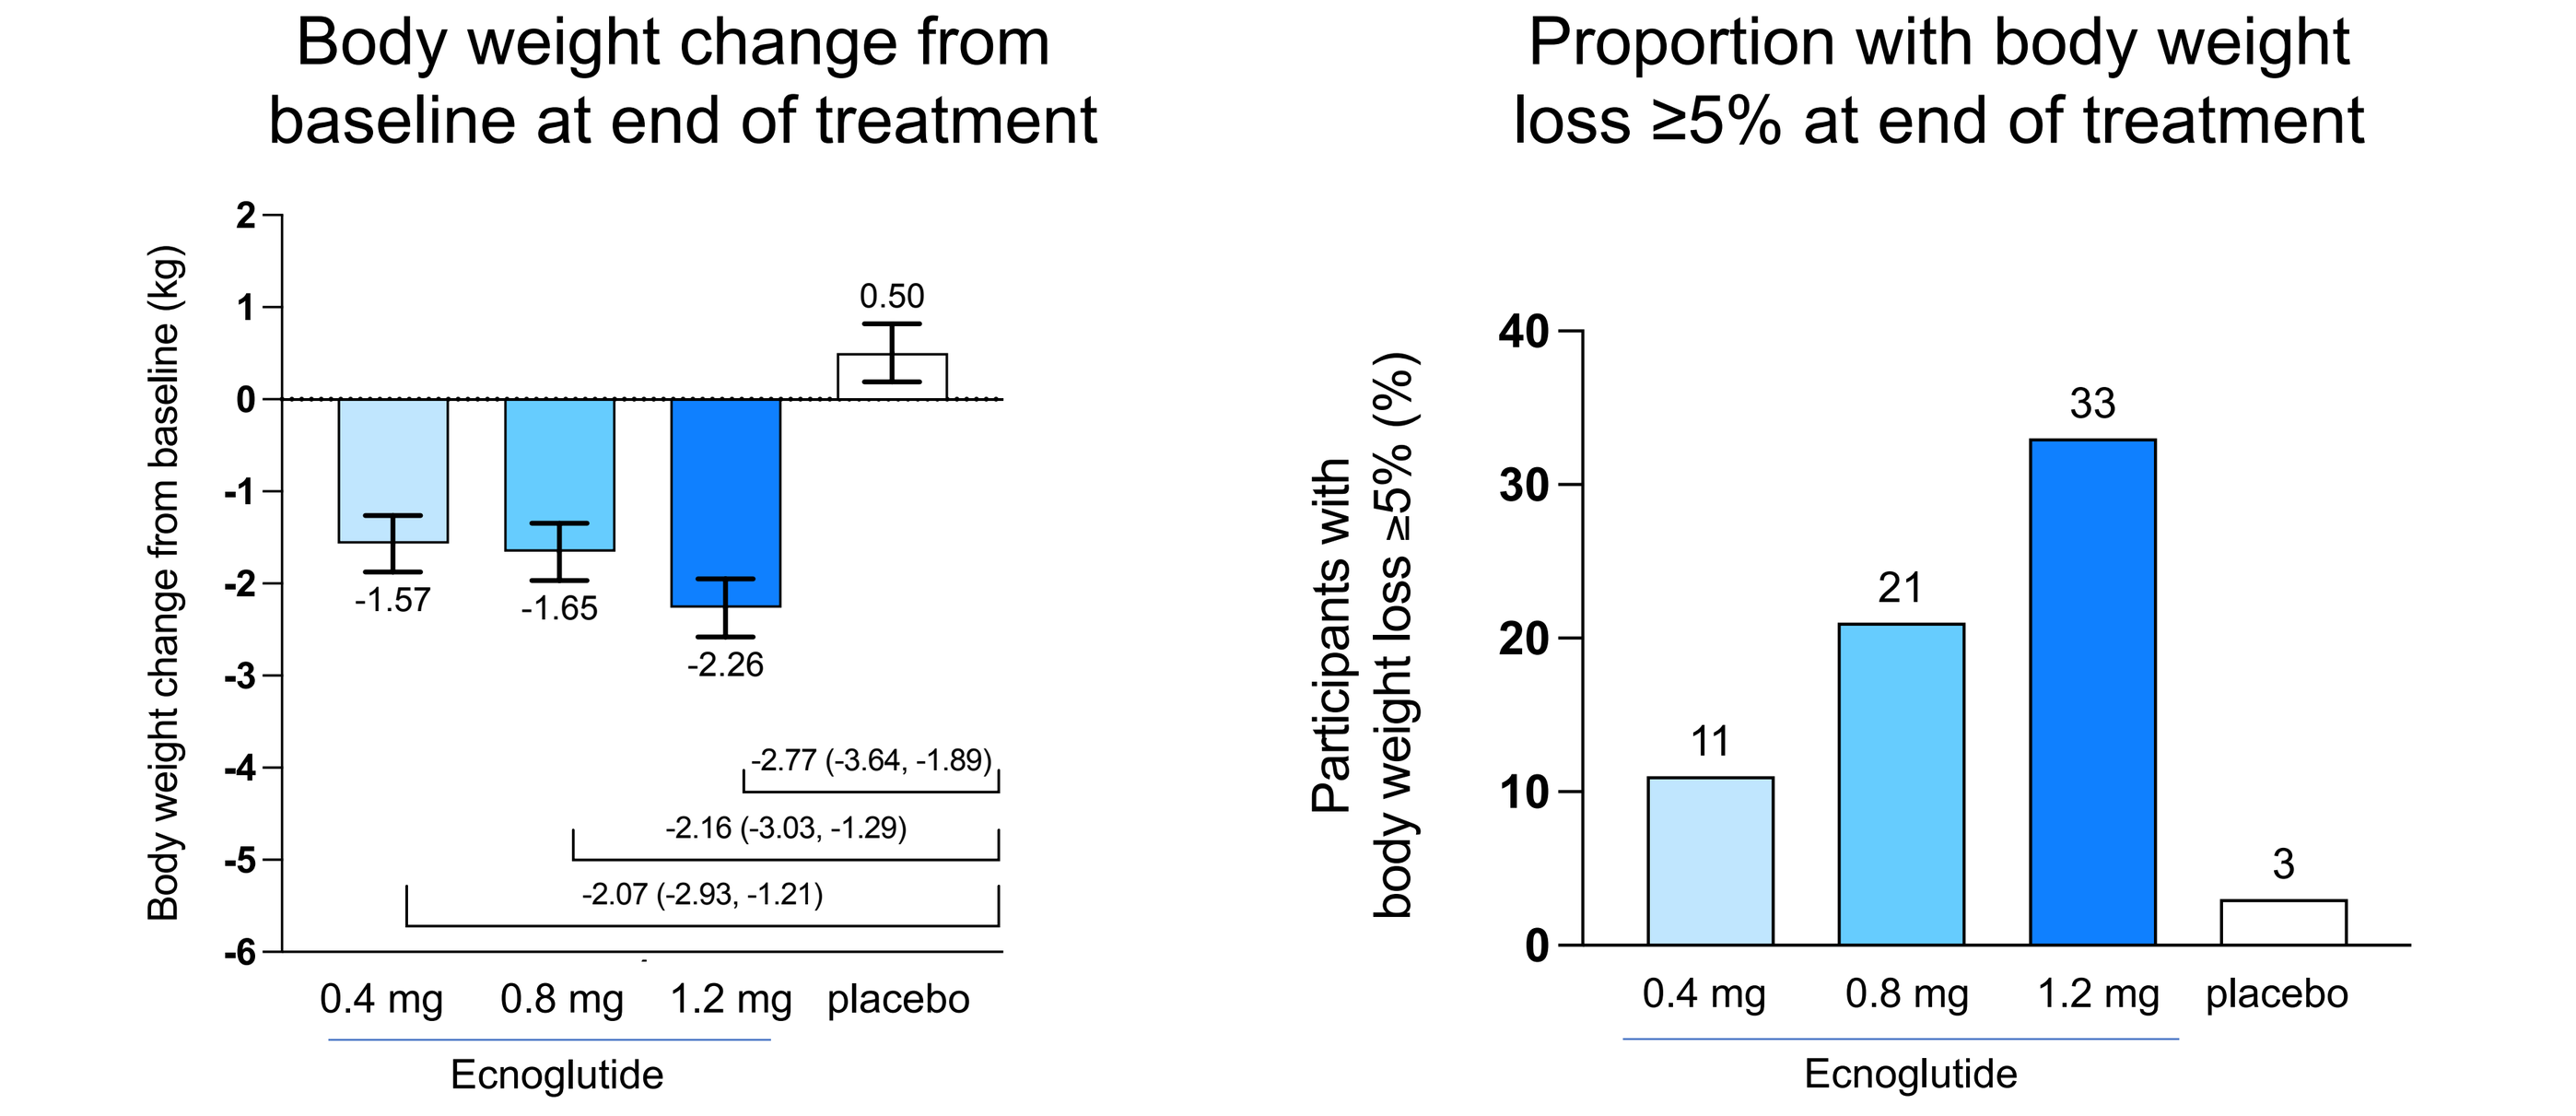
Body weight change from baseline at end of treatment
Body weight change from baseline (kg)
-1.57
-1.65
-2.26
0.50
-2.77 (-3.64, -1.89)
-2.16 (-3.03, -1.29)
-2.07 (-2.93, -1.21)
0.4 mg
0.8 mg
1.2 mg
placebo
Ecnoglutide
Proportion with body weight loss ≥5% at end of treatment
Participants with body weight loss ≥5% (%)
11
21
33
3
0.4 mg
0.8 mg
1.2 mg
placebo
Ecnoglutide

Change in body weight compared to baseline (kg) shows least squares mean, SE, mean difference from placebo and 95% confidence interval. P<0.0001 for all ecnoglutide cohorts compared to placebo.

Summary of adverse events

AEs, n (%)	Ecnoglutide 0.4 mg N=37	Ecnoglutide 0.8 mg N=36	Ecnoglutide 1.2 mg N=36	Placebo N=36
All AE	29 (78.4)	28 (77.8)	26 (72.2)	22 (61.1)
All TRAE	18 (48.6)	16 (44.4)	16 (44.4)	6 (16.7)
TEAE ≥ Grade 3	2 (5.4)	0	2 (5.6)	3 (8.3)
TRAE ≥ Grade 3	0	0	0	0
All AESI	14 (37.8)	13 (36.1)	17 (47.2)	6 (16.7)
Treatment-related AESI	14 (37.8)	12 (33.3)	15 (41.7)	2 (5.6)
TEAE leading to drug discontinuation	0	0	1 (2.8)	0
TRAE leading to drug discontinuation	0	0	1 (2.8)	0
All SAE	0	0	1 (2.8)	0
Treatment-related SAE	0	0	0	0
TEAE leading to study withdrawal	0	0	2 (5.6)	0
TRAE leading to study withdrawal	0	0	1 (2.8)	0
TEAE leading to death	0	0	0	0
TRAE leading to death	0	0	0	0

Treatment-emergent AEs with total incidence ≥5% by preferred term

TEAEs, n (%)	Ecnoglutide 0.4 mg N=37	Ecnoglutide 0.8 mg N=36	Ecnoglutide 1.2 mg N=36	Total ecnoglutide N=109	Placebo N=36	Total N=145
Participants with at least one TEAE	29 (78.4)	28 (77.8)	26 (72.2)	83 (76.1)	21 (58.3)	104 (71.7)
Diarrhea	6 (16.2)	3 (8.3)	7 (19.4)	16 (14.7)	1 (2.8)	17 (11.7)
Nausea	2 (5.4)	4 (11.1)	7 (19.4)	13 (11.9)	1 (2.8)	14 (9.7)
Hyperlipidemia	5 (13.5)	0	4 (11.1)	9 (8.3)	2 (5.6)	11 (7.6)
Upper respiratory tract infection	1 (2.7)	4 (11.1)	2 (5.6)	7 (6.4)	4 (11.1)	11 (7.6)
Constipation	2 (5.4)	3 (8.3)	3 (8.3)	8 (7.3)	2 (5.6)	10 (6.9)
Proteinuria	2 (5.4)	3 (8.3)	3 (8.3)	8 (7.3)	0	8 (5.5)

CONCLUSIONS

- Ecnoglutide led to significant HbA1c reductions of -1.81 to -2.39% from baseline to end of treatment (vs -0.55% for placebo)
- At end of treatment, up to 81% of participants receiving ecnoglutide showed HbA1c <7% (vs 19% for placebo) and up to 33% had weight loss ≥5% from baseline (vs 3% for placebo)
- HbA1c reductions for 1.2 mg ecnoglutide (-2.39%) were comparable to 1 mg semaglutide (-1.86%)[4] and 15 mg tirzepatide (-2.30%)[4]
- Ecnoglutide resulted in significant reductions in FBG and SMBG
- Ecnoglutide was generally safe and well tolerated in participants with T2DM. There were no treatment-related ≥Grade 3 or SAEs.
- Two Phase 3 studies of ecnoglutide in T2DM patients have been initiated

REFERENCES

1. International Diabetes Federation (idf.org)
2. Nadkarni et al. Prog Mol Biol Transl Sci. 2014;121:23-65
3. WHO 1999. Diagnostic Criteria for Diabetes Mellitus
4. Frias et al. N Engl J Med 2021;385:503-15

FINANCIAL DISCLOSURES

H. Qin, Q. Zheng, J. Ning, Z. Zhu, M. Guo, Y. Bu, C. L. Jones, M. Fenaux, S. Xu, and M. K. Junaidi are employees of Sciwind Biosciences.